CENTURY THERAPEUTICS

Abstract #7023

Correspondence:
krish.patel@swedish.org

Interim Results from the ELiPSE-1 Study: A Phase 1, Multicenter, Open-Label Study of CNTY-101 in Subjects with Relapsed or Refractory CD19-Positive B-Cell Malignancies

Krish Patel, Swathi Namburi, Tahir Latif, Olalekan O. Oluwole, Scott J. Cross, Gary Simmons, Chaitanya Iragavarapu, Bei Hu, Gloria Jih, Kevin Bullaughey, Poulomee A Das, Elizabeth Devlin, Kevin Flowers, Thomas Fountaine, Iphigenia Koumenis, Indu Ramachandran, Sarah Rothman, Nikolaus S. Trede, Stephanie Yee & Tamara Kay Moyo

BACKGROUND

CNTY-101 aims to deliver durable responses in R/R B-cell NHL via repeat dosing facilitated by Allo-Evasion™

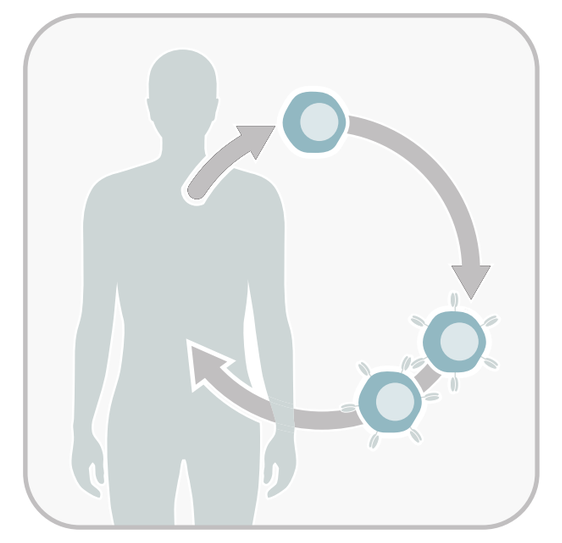

- Autologous CD19 CAR-T is curative in 40 percent of patients
- Autologous CD19 CAR-T access is limited and/or can fail in manufacturing as quality is dependent on patient-derived starting material
- Limited options and poor prognosis for patients who fail autologous CAR-T

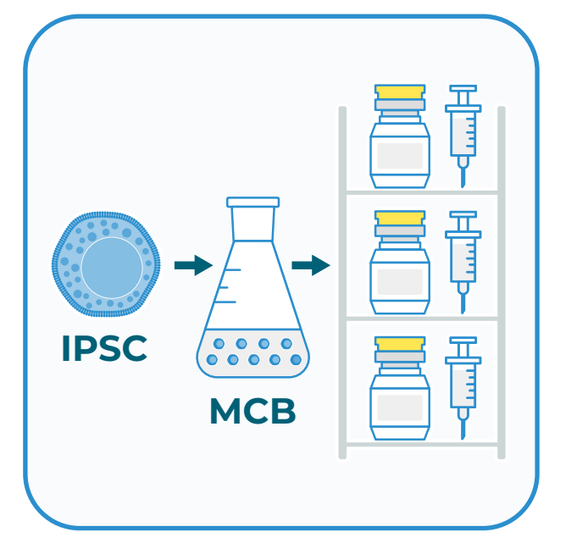
IPSC
MCB

- Off-the-shelf product offers immediate access and consistency
- Multiple doses to increase pharmacological pressure to increase durability
- Host rejection addressed by Allo-Evasion™ edits

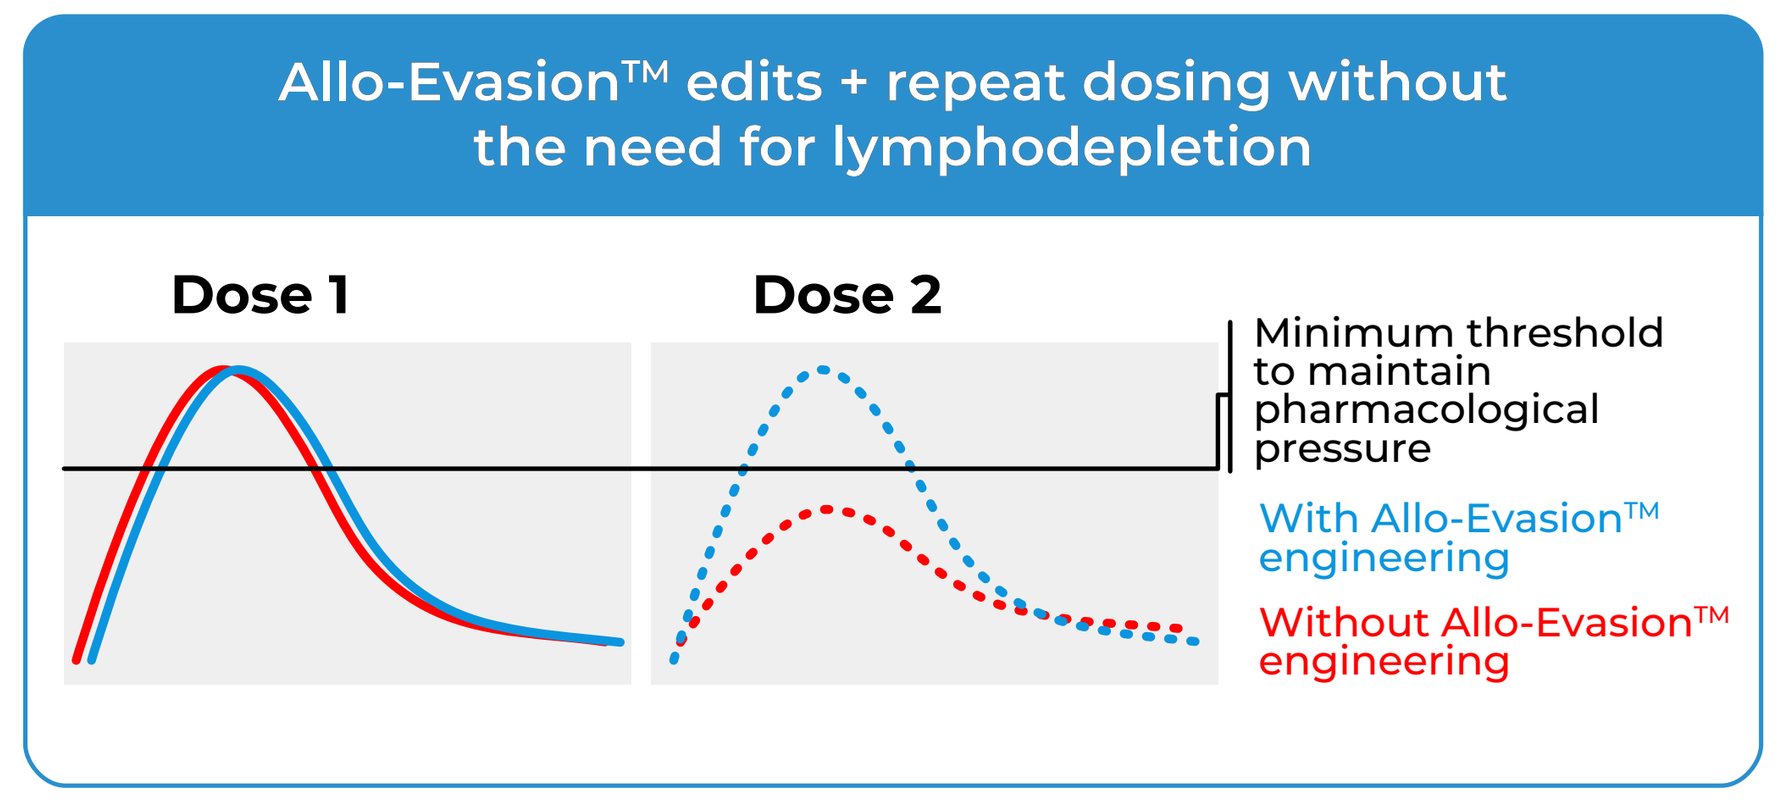
Allo-Evasion™ edits + repeat dosing without the need for lymphodepletion
Dose 1
Dose 2
Minimum threshold to maintain pharmacological pressure
With Allo-Evasion™ engineering
Without Allo-Evasion™ engineering

Aim: Extending the period of pharmacologic pressure on tumor cells

R/R: relapsed or refractory, NHL: non-Hodgkin lymphoma, CAR-T: chimeric antigen receptor T cell therapy, iPSC: induced pluripotent stem cell, MCB: master cell bank

CNTY-101 is an iPSC-derived NK cell therapy with CD19 CAR and Allo-Evasion™ edits to avoid host rejection

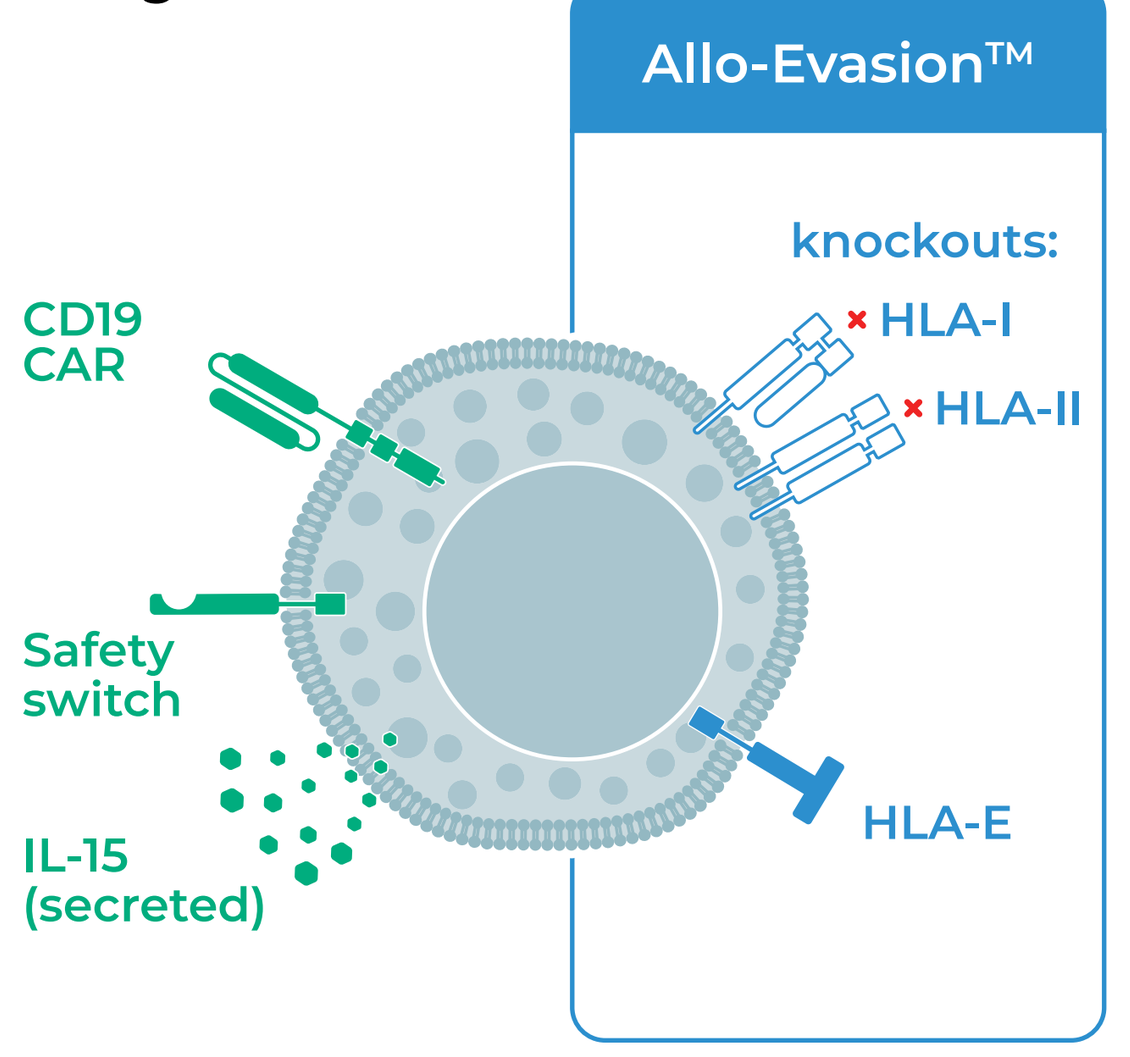
Allo-Evasion™
knockouts:
HLA-I
HLA-II
HLA-E
CD19 CAR
Safety switch
IL-15 (secreted)

CNTY-101 is an allogeneic anti-CD19 iPSC-derived NK cell therapy

Century's technology for multiple step editing using CRISPR in iPSC allowed 6 edits (2 KO, 4 KI) to enable multiple functions

- Avoid host T cell rejection: Knockout of beta-2 microglobulin (b2M) and HLA Class II Transactivator (CIITA) to eliminate HLA-I & HLA-II
- Avoid host NK cell rejection: Knock-in of HLA-E
- Tumor elimination: CD19 CAR with FMC63 binder
- Safety switch: truncated Epidermal Growth Factor Receptor (EGFRt) containing Cetuximab binding epitope
- Secreted IL-15: to enhance cell persistence and modulate the tumor microenvironment

NK: natural killer, FMC63: CD19 binding portion of CAR, HLA: human leukocyte antigen, KO: knockout, KI: knock-in

STUDY DESIGN

CNTY-101: ELiPSE-1 (NCT05336409) Phase 1 BOIN Design

Patients with CD19+ aggressive and high-risk indolent R/R B-NHL
- DLBCL, HGBL, MCL, PMBCL, FL3B, HRFL, MZL
- ≥ 2 prior lines of therapy
- Prior CD19-targeted cell therapy allowed

Part 1 - Dose escalation
Schedule A: single dose
Schedule B: 1 dose per week x 3 weeks
Part 2 - Dose expansion

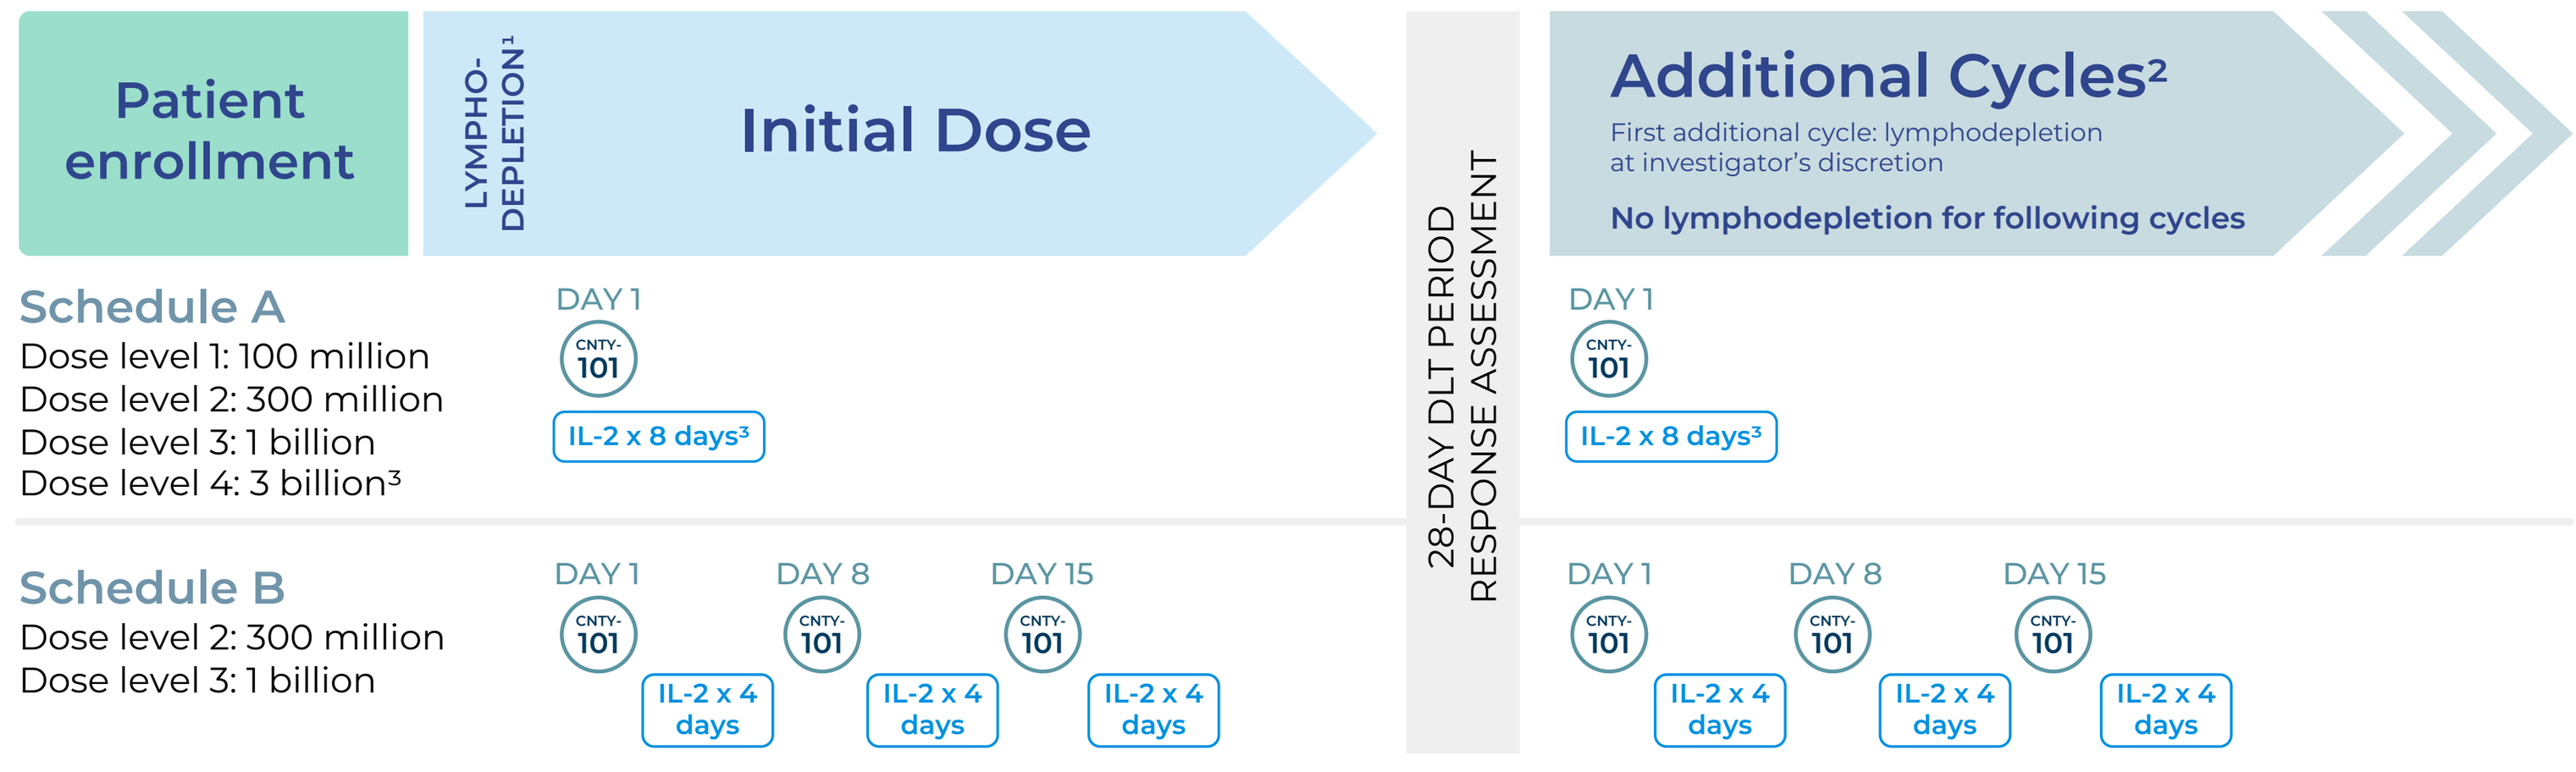
Patient enrollment
LYMPHO-DEPLETION¹
Initial Dose
28-DAY DLT PERIOD RESPONSE ASSESSMENT
Additional Cycles²
First additional cycle: lymphodepletion at investigator's discretion
No lymphodepletion for following cycles
Schedule A
Dose level 1: 100 million
Dose level 2: 300 million
Dose level 3: 1 billion
Dose level 4: 3 billion³
DAY 1
CNTY-101
IL-2 x 8 days³
Schedule B
Dose level 2: 300 million
Dose level 3: 1 billion
DAY 1
DAY 8
DAY 15
IL-2 x 4 days

¹Standard lymphodepletion regimen: fludarabine (30 mg/m²/d) and cyclophosphamide IV (300 mg/m²/d) for 3 days
²Subjects who are assessed as stable disease or better may receive additional cycles of CNTY-101
³Subjects at DL4A did not receive IL-2 on the day of CNTY-101 infusion but did receive daily for 7 days

BOIN: Bayesian Optimal Interval, DLT: dose limiting toxicity; IL-2: Interleukin-2 (dose: 3e6 IU; subcutaneous)

RESULTS

Baseline disease characteristics

Heavily pre-treated R/R B-NHL patients treated across 7 sites

	N=12 safety evaluable¹
Median Age (range, years)	70 (60-76)
Male, n (%)	9 (75)
NHL subtype, n (%)	
DLBCL	7 (58)
HRFL	1 (8)
MCL	2 (17)
MZL	2 (17)
Prior therapies, median (range)	4 (2-5)
Response to last line of treatment	
Relapsed	3 (25)
Refractory	9 (75)
Received prior autologous CAR-T	3(25)
If no, why	
Manufacturing fail	1
Not eligible	3
Not willing to wait	4²
Financial or reimbursement constraints	1

¹As of 27 March 2024 data cutoff date, data collection ongoing ²One subject received allogeneic CAR-T

Treatment Emergent Adverse Events (TEAEs)

Manageable safety profile to date; No DLTs or GVHD observed
8/12 subjects received at least one cycle of CNTY-101 in an outpatient setting

	Any Grade n (%)	Grade 3+ n(%)	Related Gr 3+ n(%)
# of subjects with TEAEs	12 (100)	11 (92)	2 (17)
Neutropenia	10 (83)	10 (83)	1 (8)
Nausea	6 (50)	1 (8)	
Anaemia	6 (50)	4 (33)	1 (8)
Thrombocytopenia	6 (50)	1 (8)	
Hypotension	5 (42)	1 (8)	
Constipation	4 (33)		
Febrile neutropenia	4 (33)	4 (33)	1 (8)
Fatigue	4 (33)		
Pyrexia	4 (33)		
Chills	4 (33)		
Cytokine release syndrome	4 (33)		
Diarrhoea	3 (25)		
Hyperhidrosis	3 (25)		
Injection site reaction	3 (25)		
Muscular weakness	3 (25)	2 (17)	
Vomiting	3 (25)		

- Only TEAEs in at least n=3 subjects of any grade are shown
- 12 subjects evaluable for safety across 4 dose levels and two dosing schedules¹

EVENT		n=12
CRS, n(%)		
	Any Grade	4 (33)
	Grade 1	2 (17)
	Grade 2	2 (17)
Median days to onset, days (range)		0 (0-6)
	Fever >24 hrs	1 (8)
	Hypotension	1 (8)
	Hypoxia	1 (8)

- Only one Gr 1 CRS event in n=1 subject was associated with a fever that lasted more than 24 hrs
- Hypotension (n=1) and hypoxia (n=1) lasted less than 24 hrs

EVENT		n=12
ICANS, n(%)		
	Any Grade	1 (8)
	Grade 1	1 (8)
Days to onset		6
	Fatigue	1 (8)
	Aphasia	1 (8)

ICANS duration was 1 day

¹As of 27 March 2024 data cutoff date, data collection ongoing. Grading based on CTCAE v5; CRS and ICANS graded per ASTCT.

CNTY-101 preliminary efficacy

30% CRR; 40% ORR

SCHEDULE A (single infusion)

Dose	Subject	Subtype	Prior Lines of Therapy	Prior CAR-T
1 billion cells	8	DLBCL	2	N
	9	MCL	4	N
300 million cells	5	MZL	4	N
	6	DLBCL	4	Y
	7	DLBCL	5	Y
100 million cells	1	HRFL	4	N
	2	DLBCL	4	Y²
	3	DLBCL	2	N
	4	DLBCL	3	N

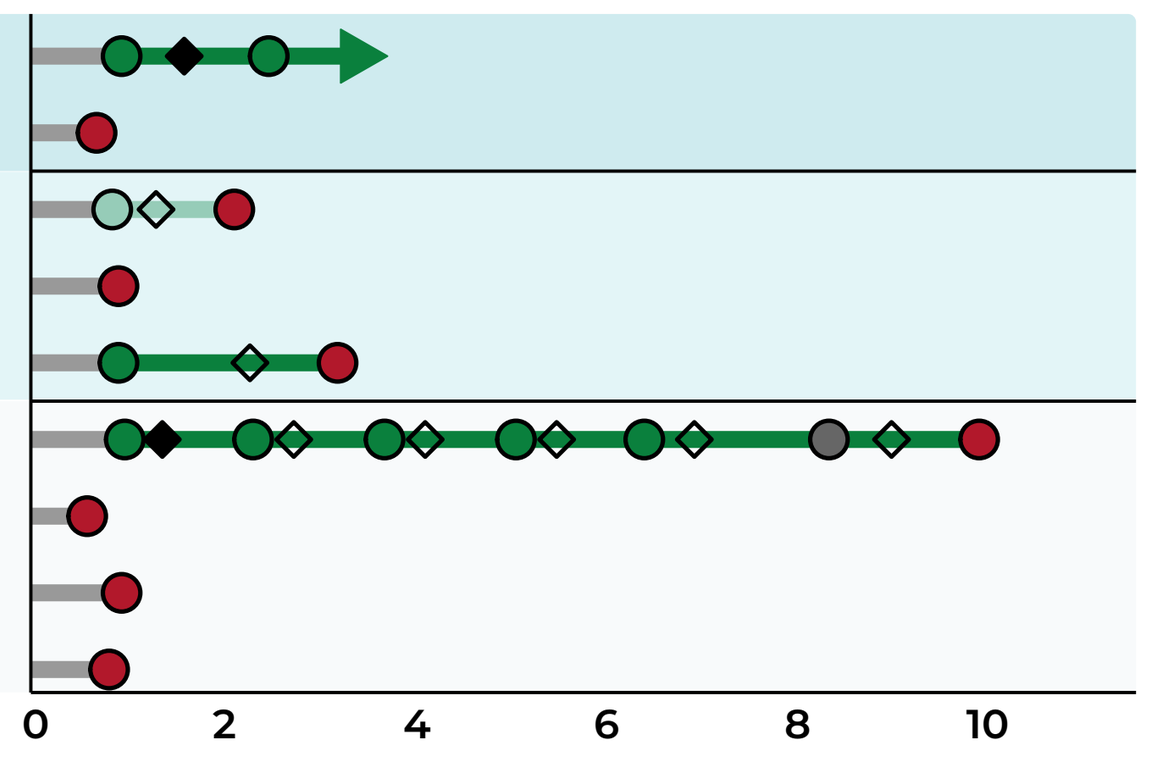
0
2
4
6
8
10
Time from first CNTY-101 infusion (months)

SCHEDULE B (3 weekly infusions)

Dose	Subject	Subtype	Prior Lines of Therapy	Prior CAR-T
300 million cells	10	MZL	4	N

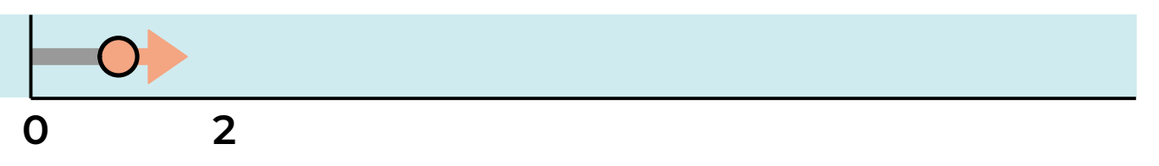
0
2
Time from first CNTY-101 infusion (months)

10 subjects evaluable for efficacy across 3 dose levels and two dosing schedules¹

CR, PR, SD, PD, NE
◆ Dose (with LDC)
◇ Dose (no LDC)
HRFL: high risk follicular lymphoma
DLBCL: diffuse large B cell lymphoma
MZL: marginal zone lymphoma
MCL: mantle cell lymphoma

¹As of 27 March 2024 data cutoff date, data collection ongoing, efficacy based on Lugano criteria ²Subject received prior allogeneic CAR-T

CRR: complete response rate, ORR: overall response rate

CNTY-101 rapidly traffics out of circulation and persists in extravascular space

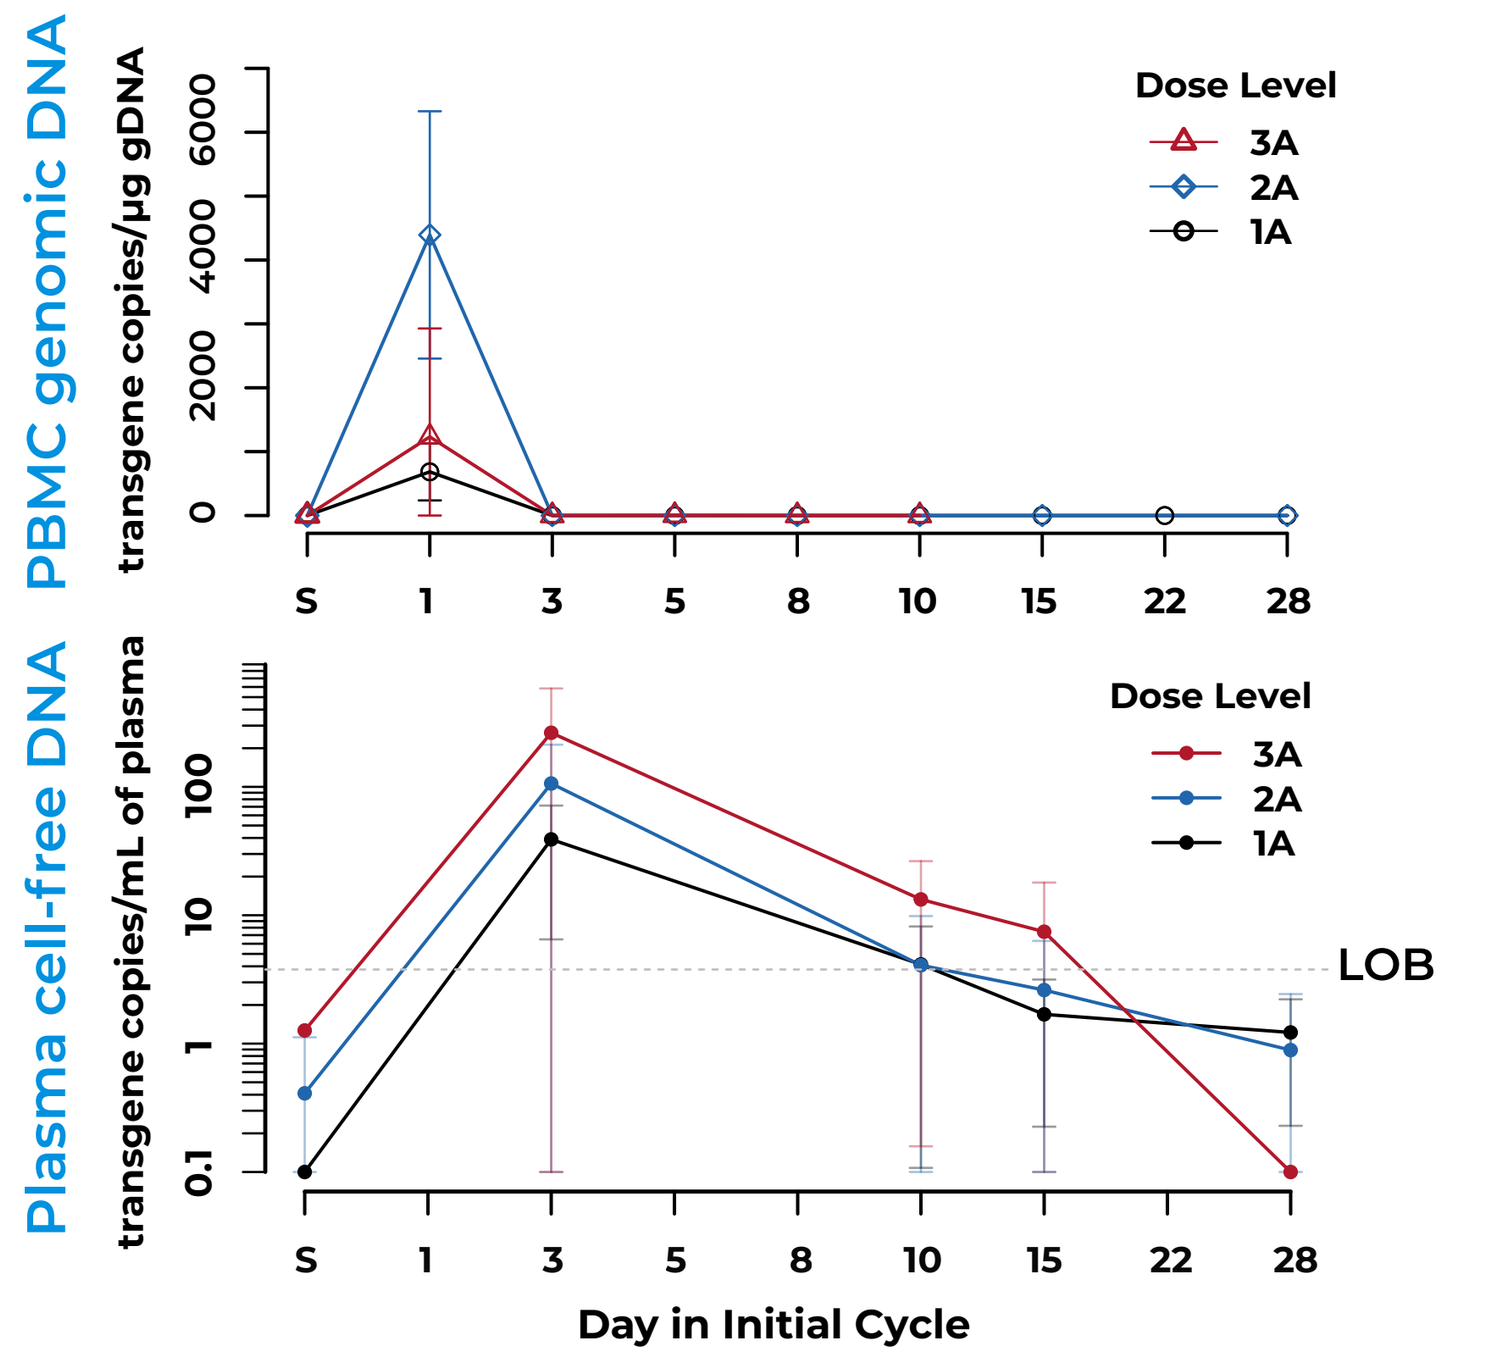
PBMC genomic DNA
transgene copies/µg gDNA
Plasma cell-free DNA
transgene copies/mL of plasma
Dose Level
3A
2A
1A
LOB
Day in Initial Cycle

CNTY-101 has limited duration in circulation; maximum concentration is observed at Day 1 post-infusion

Transgene copies per µg genomic DNA were determined using ddPCR with primers targeting transgene and RPP30. Data is shown for initial cycle across subjects at each dose level; mean ± SD. LLOQ is 5 copies.

CNTY-101 persistence is detected via cell-free DNA¹ on Day 3 and beyond

Transgene copies per mL of plasma were determined using ddPCR. Data is shown for initial cycle across subjects at each dose level; mean ± SD (error bar: low values are truncated at 0.1). Positivity values are determined to be significantly above LOB using two sample Poisson test, $p < 0.05$.

¹Cell-free DNA has short half-life in circulation, ranging from minutes to hours (Khier and Lohan, Future Science 2018)

S: screen, ddPCR: droplet digital PCR, RPP30: Ribonuclease P protein subunit p30, LOB: limit of blank, SD: standard deviation, LLOQ: lower limit of quantification

Patient 1 (Schedule A 100 million) Case Study - No signs of allo-rejection observed after multiple treatment cycles without LDC

Detection of CNTY-101 cell-free DNA (cfDNA)

Cycle	Initial Cycle	AC1	AC2	AC3	AC4	AC5	AC6
DAY3	not collected	30 [+]	16 [+]	24 [+]	21 [+]	30 [+]	19 [+]
DAY10	not collected	3 [+]	6 [+]	3 [+]	0 [–]	not collected	not collected
DAY15	not collected	2 [–]	not collected	2 [–]	1 [–]	not collected	2 [–]
DAY28	6 [+]	2 [–]	6 [+]	2 [–]	2 [–]	1 [–]	3 [+]

LDC / No LDC; p value: < 1e-10, 1e-8, 1e-6, 1e-4, 1e-2, 1

Consistent detection of CNTY-101 cfDNA on Day 3 across all cycles delivered with or without LDC

Transgene copies detected in 1 mL of plasma is indicated. Detectable signal [+] was determined to be significantly above negative controls using two sample Poisson test, $p < 0.05$.

AC: additional cycle, ND: not detected

Antibody-dependent cellular cytotoxicity (ADCC)

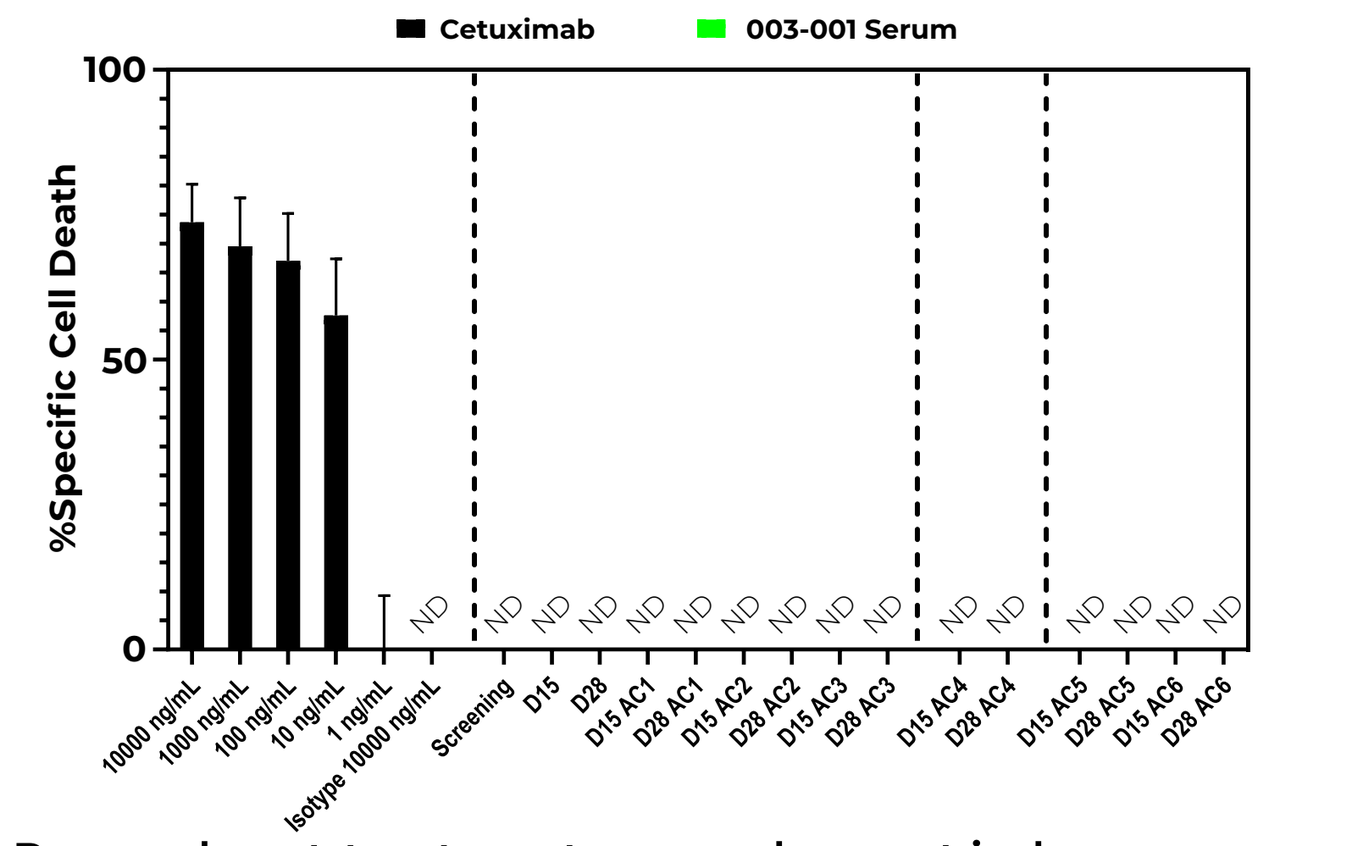
Cetuximab
003-001 Serum
%Specific Cell Death
ND

Pre- and post-treatment serum does not induce ADCC-mediated cell death of CNTY-101

ADCC-mediated cell death of CNTY-101 targets was measured by pre-incubation with monoclonal control antibodies or patient serum, followed by the addition of NK-92-CD16-V158 effectors. Controls were pooled across plates; individual plate runs are separated by dashed vertical bars. Results are represented as mean +/- SD.

Complement-dependent cytotoxicity (CDC)

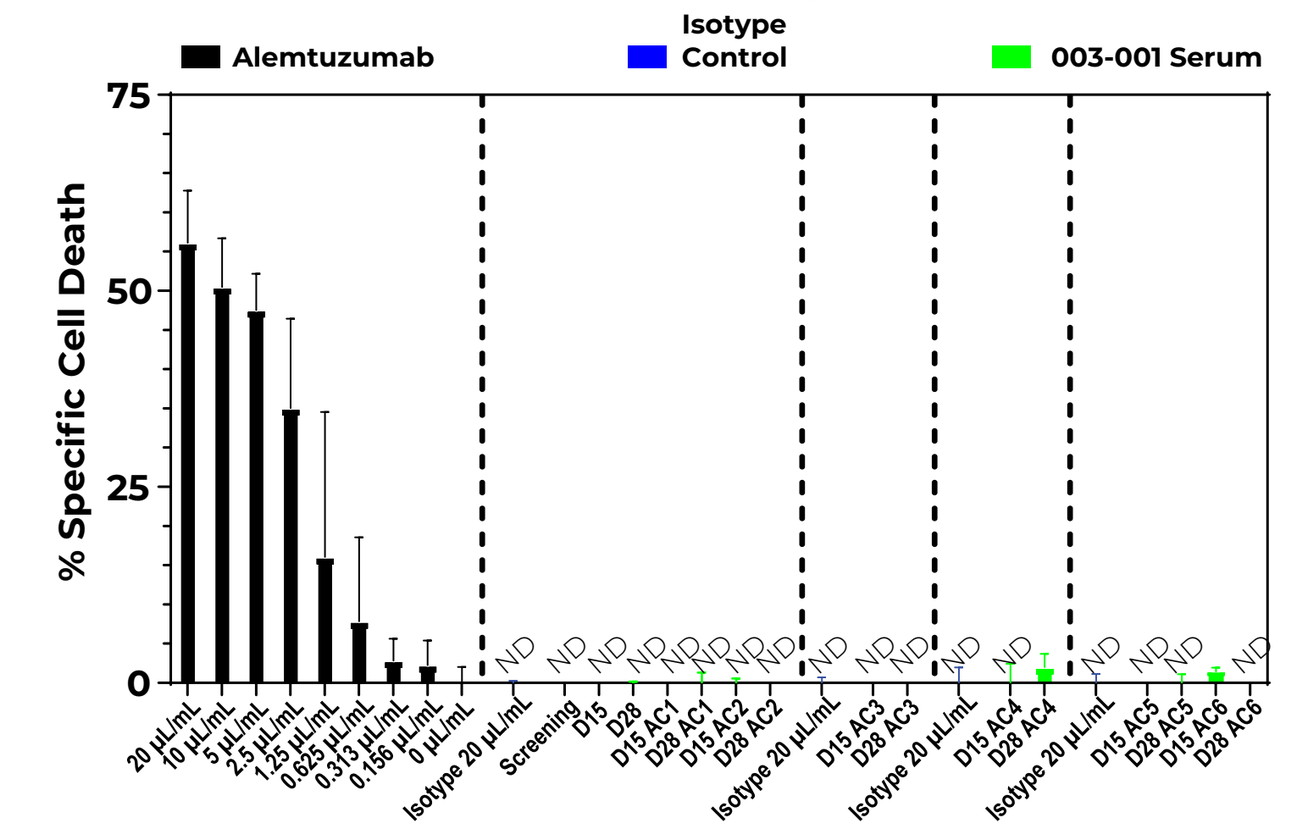
Alemtuzumab
Isotype Control
003-001 Serum
% Specific Cell Death
ND

Pre- and post-treatment serum does not induce CDC-mediated cell death of CNTY-101

CDC-mediated cell death of CNTY-101 targets was measured by pre-incubation with control monoclonal antibodies or patient serum, followed by addition of human complement. Positive controls were pooled across plates; individual plate runs are separated by dashed vertical bars. Results are represented as mean +/- SD.

SUMMARY

- CNTY-101 is the first iPSC-derived NK cell therapy engineered with six precision gene edits including Allo-evasion™ edits.
- Infusions of up to 1 billion CNTY-101 cells per cycle (as single 1 billion or 3 x 300 million) in patients with R/R NHL demonstrate a manageable safety profile, including in the outpatient setting, with no GvHD or DLTs to date.
- Preliminary efficacy shows 30% CRR rate (n=10) in heavily pretreated, mostly refractory patients with predominantly aggressive or high-risk histologies.
- Utilizing a novel cell-free DNA method for detecting total body PK of CNTY-101, we show that CNTY-101 rapidly traffics out of circulation and persists in the extravascular space. AUC is trending to increase with dose.
- Patient 1 received five cycles of CNTY-101 (100 million) without LDC and allo-rejection was not observed.
- Overall, in 3 out of 4 patients who received CNTY-101 in cycles without LDC, positive detection of CNTY-101 was observed on Day 3 at minimum.

Current study status: Enrollment is ongoing; in dose escalation phase
Enrolling at dose level 3B (1 billion x 3 doses) and 4A (3 billion x 1 dose)

Clinical data as of 27 March, 2024; translational data as of 1 May, 2024

GvHD: graft versus host disease, DLT: dose limiting toxicities, PK: pharmacokinetics, AUC: area under the curve

Acknowledgements: We thank Thomas Brigman, Sam Kelly, Kile McFadden and Brenda Salantes for their contributions.